809

Response of Metabolic Phenotype to Chemotherapy Agents in a Simulated Tumor Microenvironment

David L. Hoffman and Julie M. Rumble
Cayman Chemical, Ann Arbor, Michigan

Abstract

Chemotherapeutic agents (CAs), while standard-of-care for treating numerous types of cancer, are by nature, poisonous. Studies have shown that treatment with certain CAs, such as irinotecan, leads to chemotherapy-associated steatohepatitis (CASH) resulting from the inhibition of mitochondrial fatty acid oxidation (FAO). While inhibition of FAO may have harmful effects on the liver, it may have a different effect in the tumor microenvironment (TME). Within the TME exists a unique metabolic environment, consisting of low [O_2], low pH, and high concentrations of fatty acids. Occupying the TME are immunosuppressive myeloid-derived suppressor cells (MDSCs), which aid in tumor growth by suppressing T cells, which would normally attack the tumor. It is thought that the MDSCs localized to the TME undergo an alteration in metabolic phenotype, allowing them to utilize the high concentrations of fatty acids found in the TME as their primary carbon source, becoming reliant on FAO. It is therefore hypothesized that normoxic MDSCs will alter their metabolic phenotype in response to a simulated TME to favor FAO. This altered metabolic phenotype will sensitize MDSCs to inhibition of FAO by SN-38, the active metabolite of irinotecan. The response of metabolic phenotype to hypoxic conditions will also be examined in HCT116 colorectal carcinoma cells.

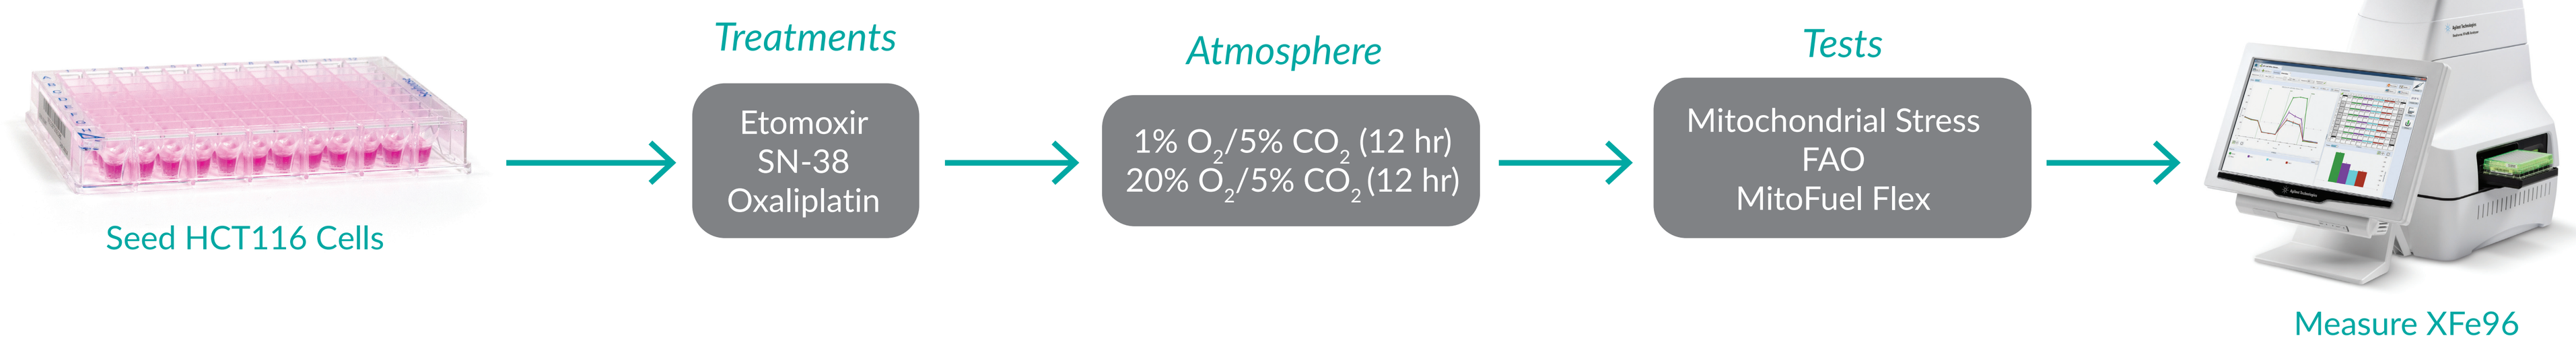
Treatments
Atmosphere
Tests
Seed HCT116 Cells
Etomoxir
SN-38
Oxaliplatin
1% O_2/5% CO_2 (12 hr)
20% O_2/5% CO_2 (12 hr)
Mitochondrial Stress
FAO
MitoFuel Flex
Measure XFe96

Figure 1. HCT116 cells were seeded at 1 x 10^5 cells/well and allowed to settle for 1 hour before treatment with compounds. Cells were cultured overnight in DMEM supplemented with 10% FBS, 1 mM glutamine, and pen/strep at 37°C/5% CO_2 at ambient O_2 or 1% O_2. The following morning, complete DMEM was removed and replaced with XF DMEM supplemented with 10 mM glucose, 1 mM glutamine, and 1 mM pyruvate (Agilent Technologies). Cells were degassed and imaged using the Cytation™ 5 Imaging Multimode Plate Reader (BioTek Instruments) before performing all experiments. For FAO experiments see figure legend for **Figure 3**.

Mitochondrial Stress Test

21% O_2 *1% O_2*

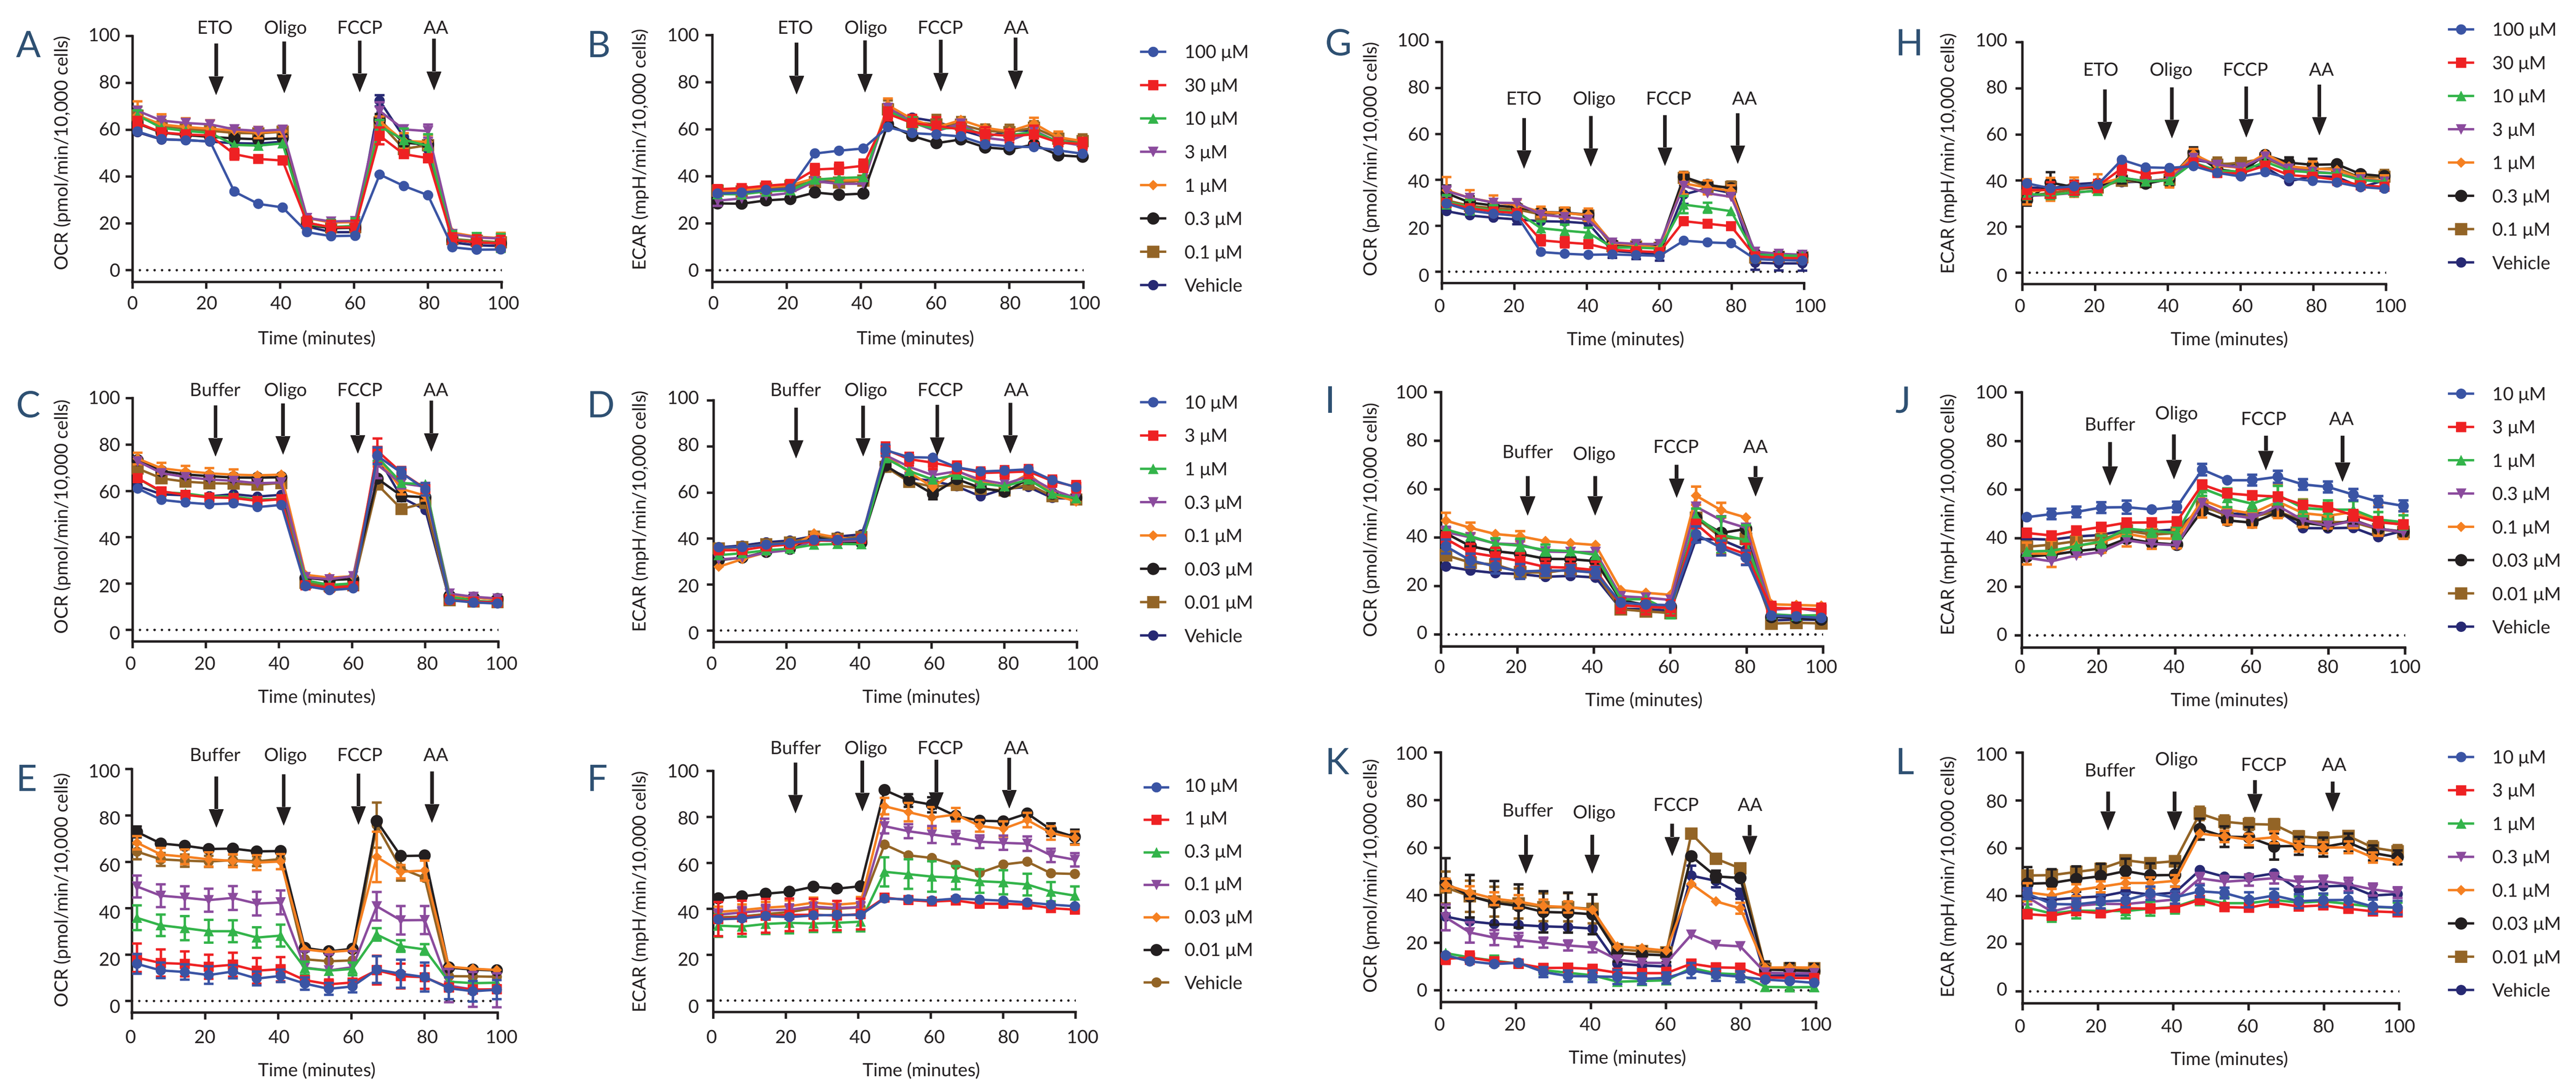

Figure 2. HCT116 cells cultured at 21% O_2 OCR (**A.**) and ECAR (**B.**) response to acute treatment with etomoxir, prolonged treatment with SN-38 (**C.** and **D.**), and prolonged treatment with oxaliplatin (**E.** and **F.**). HCT116 cells cultured at 1% O_2 response to acute treatment with etomoxir (**G.** and **H.**), prolonged treatment with SN-38 (**I.** and **J.**), and prolonged treatment with oxaliplatin (**K.** and **L.**). For all experiments, final concentrations of inhibitors: oligomycin (1 µg/ml), FCCP (1 µM), antimycin A/Hoechst (10 µM/100 µM).

To compare the effects of CA to fatty oxidation inhibitors in tumor cells, HCT116 cells were pretreated with SN-38 (which has previously been shown to inhibit FAO), oxaliplatin, and the CPT1 inhibitor etomoxir (acutely) and cultured at 21% and 1% O_2. Following treatment, a mitochondrial stress test, utilizing oligomycin (complex V inhibitor), FCCP (uncoupler, maximizing OCR), and antimycin A (inhibits mitochondrial OCR) was performed in order to determine the effect of each condition on the cell's mitochondrial phenotype. Treatment with the CPT1 inhibitor etomoxir (**Figure 2A**) at 21% O_2 shows inhibition of OCR at 30 µM and above. However, since the oligomycin-induced OCR does not change when compared to vehicle, this suggests a reduction in metabolite availability to the electron transport chain. Indeed, an increase in ECAR is observed (**Figure 2B**) upon addition of etomoxir, showing that glycolysis increases due to the inhibition of FAO. Prolonged treatment with SN-38 at concentrations between 0.03-1 µM appears to increase basal OCR without causing uncoupling (**Figure 2C**). The response of glycolysis (ECAR) to the ATP synthase inhibitor oligomycin does not appear to be affected by SN-38 (**Figure 2D**). Treatment with oxaliplatin (**Figure 2E**) at concentrations greater than 0.3 µM inhibits both OCR and glycolysis (**Figures 2E and F**), indicating cellular toxicity. Cells cultured at 1% O_2 overall had a decreased OCR, and a slightly higher ECAR, as expected. At 1% O_2, cells appeared more sensitive to etomoxir when compared to those cultured at 21% (**Figures 2G and H**). Treatment with SN-38 (**Figures 2I and J**) showed a similar response to that at 21% O_2 as did oxaliplatin (**Figures 2K and L**).

FAO Stress Test

21% O_2 *1% O_2*

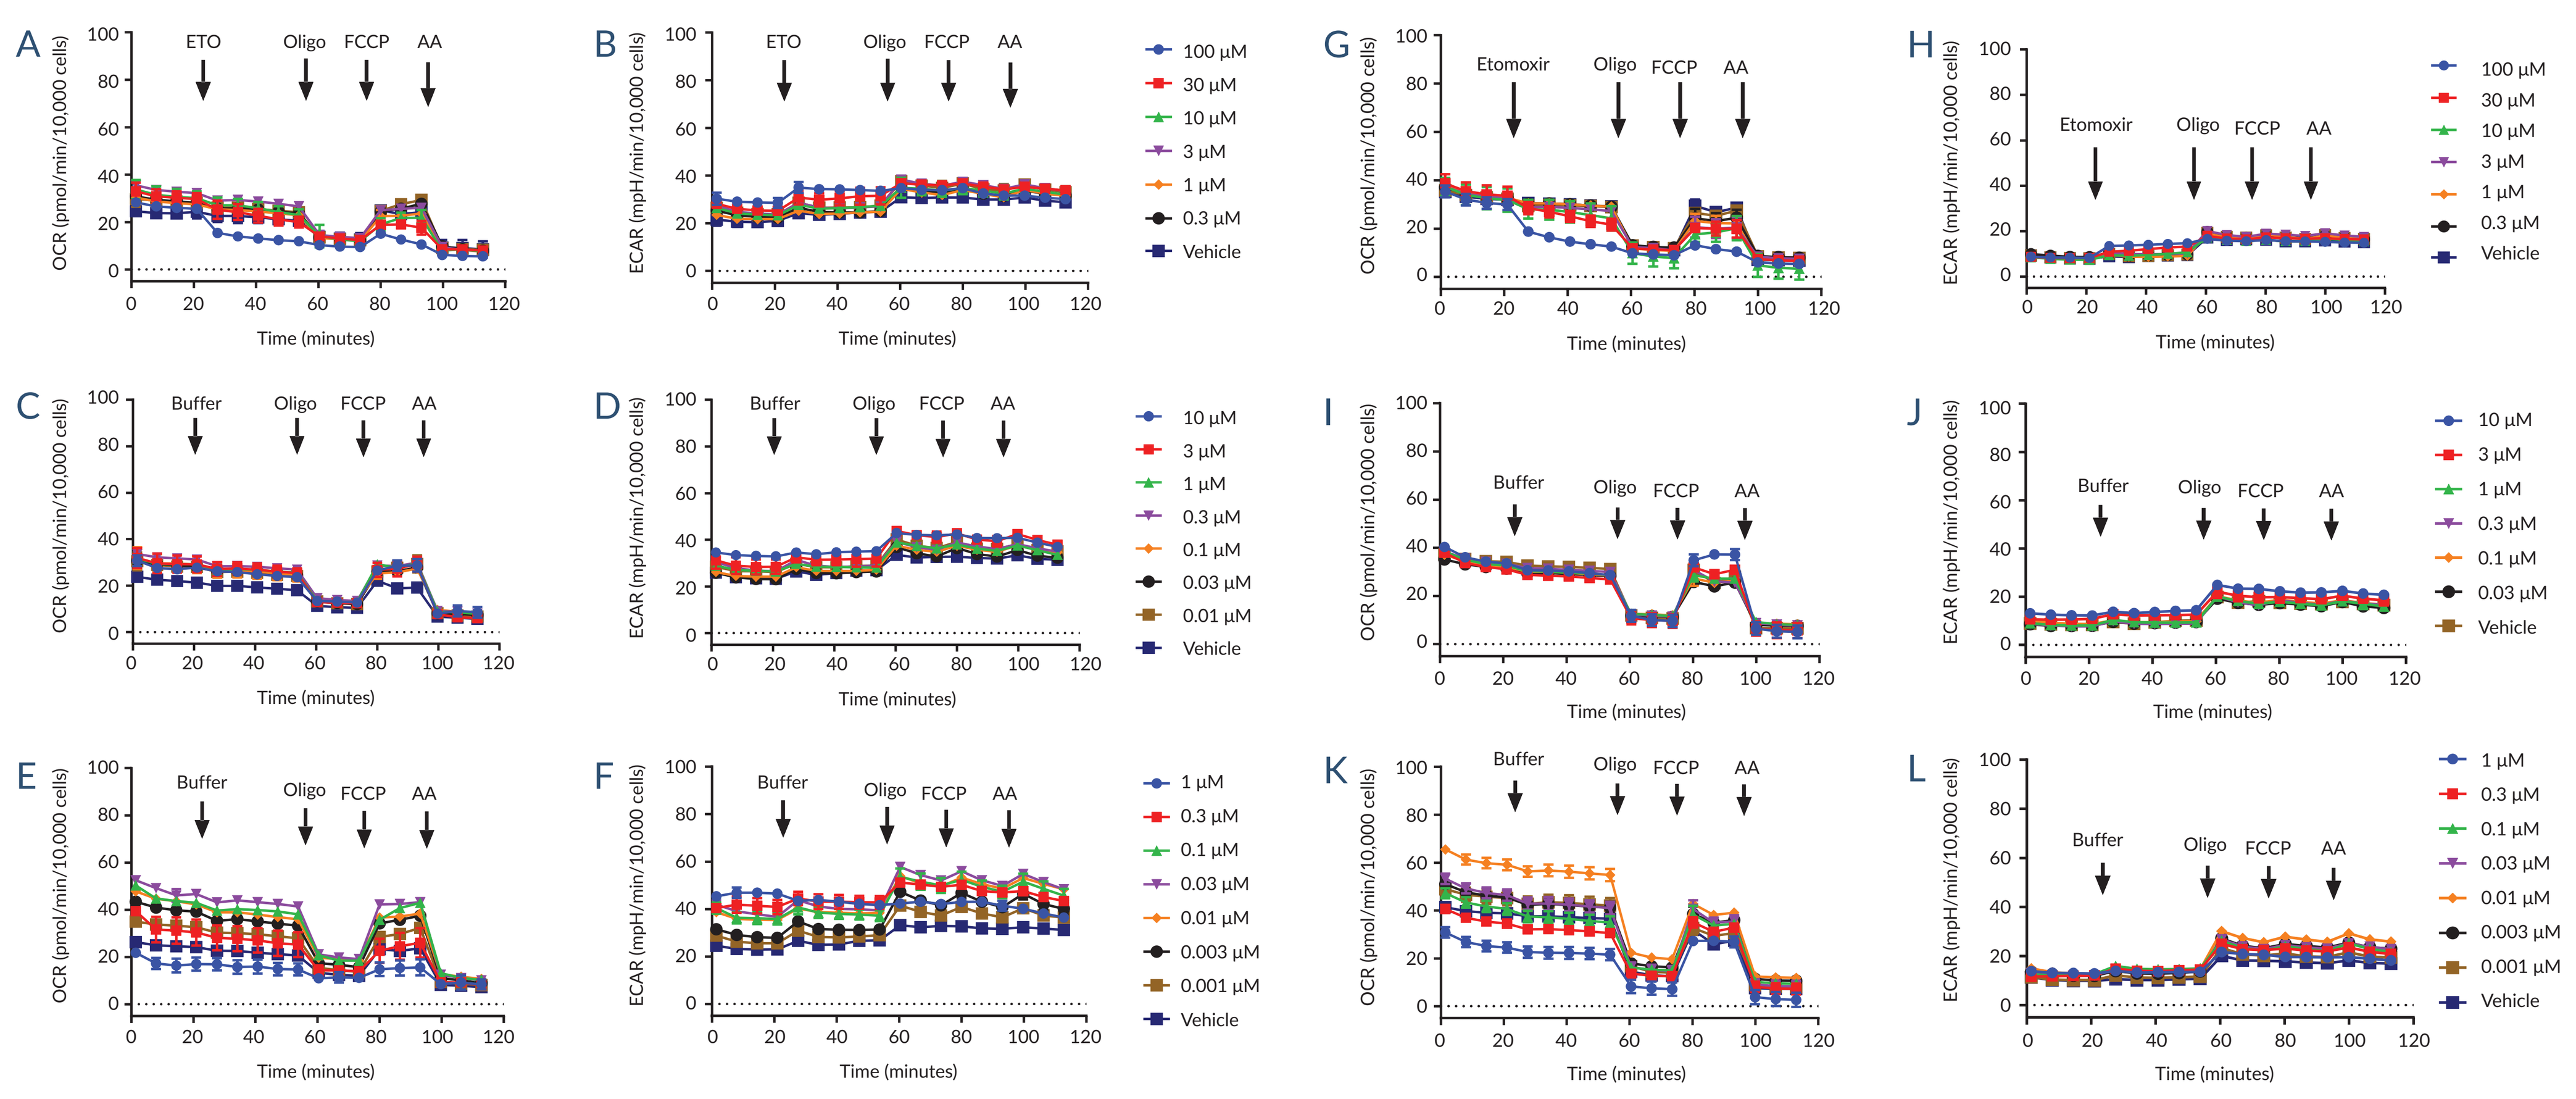

Figure 3. In order to maximize the effects of SN-38 and oxaliplatin on FAO, cells were cultured for 24 hours in low nutrient/low serum media in the presence of carnitine, maximizing the metabolic dependence on exogenous fatty acids, and thereby upregulating FAO. CAs were added for the last 12 hours of the culture period and cultured at 21% or 1% O_2. Following the treatment period, cells were placed in a modified low-glucose Krebs-Henseleit buffer containing carnitine and supplemented with BSA:palmitate as a source of fatty acid. The mitochondrial stress test was then performed to measure mitochondrial function when utilizing FAO. Concentrations of inhibitors were consistent with those in **Figure 2**.

Under normoxic conditions, SN-38 had minimal effects on OCR except for a slight increase over vehicle with the addition of FCCP (**Figure 3C**). No differences in ECAR were immediately apparent with SN-38 treatment (**Figure 3D**). Treatment with oxaliplatin exhibited a similar profile as with the standard mitochondrial stress test, suggesting cellular toxicity at the concentrations tested (**Figure 3E**). This effect was also mirrored in ECAR (**Figure 3F**). As with the standard mitochondrial stress test, etomoxir treatment also results in a similar profile, showing inhibition at concentrations of 100 and 30 µM (**Figure 3A**) with ECAR following suit. This was also observed at 1% O_2, but only with lower OCR values and higher ECARs as is expected at lower O_2 concentrations. Given the similarities between the standard and FAO stress tests, these data suggest that FAO is an important metabolic pathway for HCT116 cells at 21% O_2. Based on the data collected, it was determined that oxaliplatin did not affect FAO, but rather resulted in cellular toxicity. Because of this, SN-38 (30 nM) was the primary focus of future experiments. In order to examine this further, a mitochondrial fuel flex test was performed under each condition in order to determine cellular flex for different metabolites under each condition.

Mitochondrial Fuel Flex Test

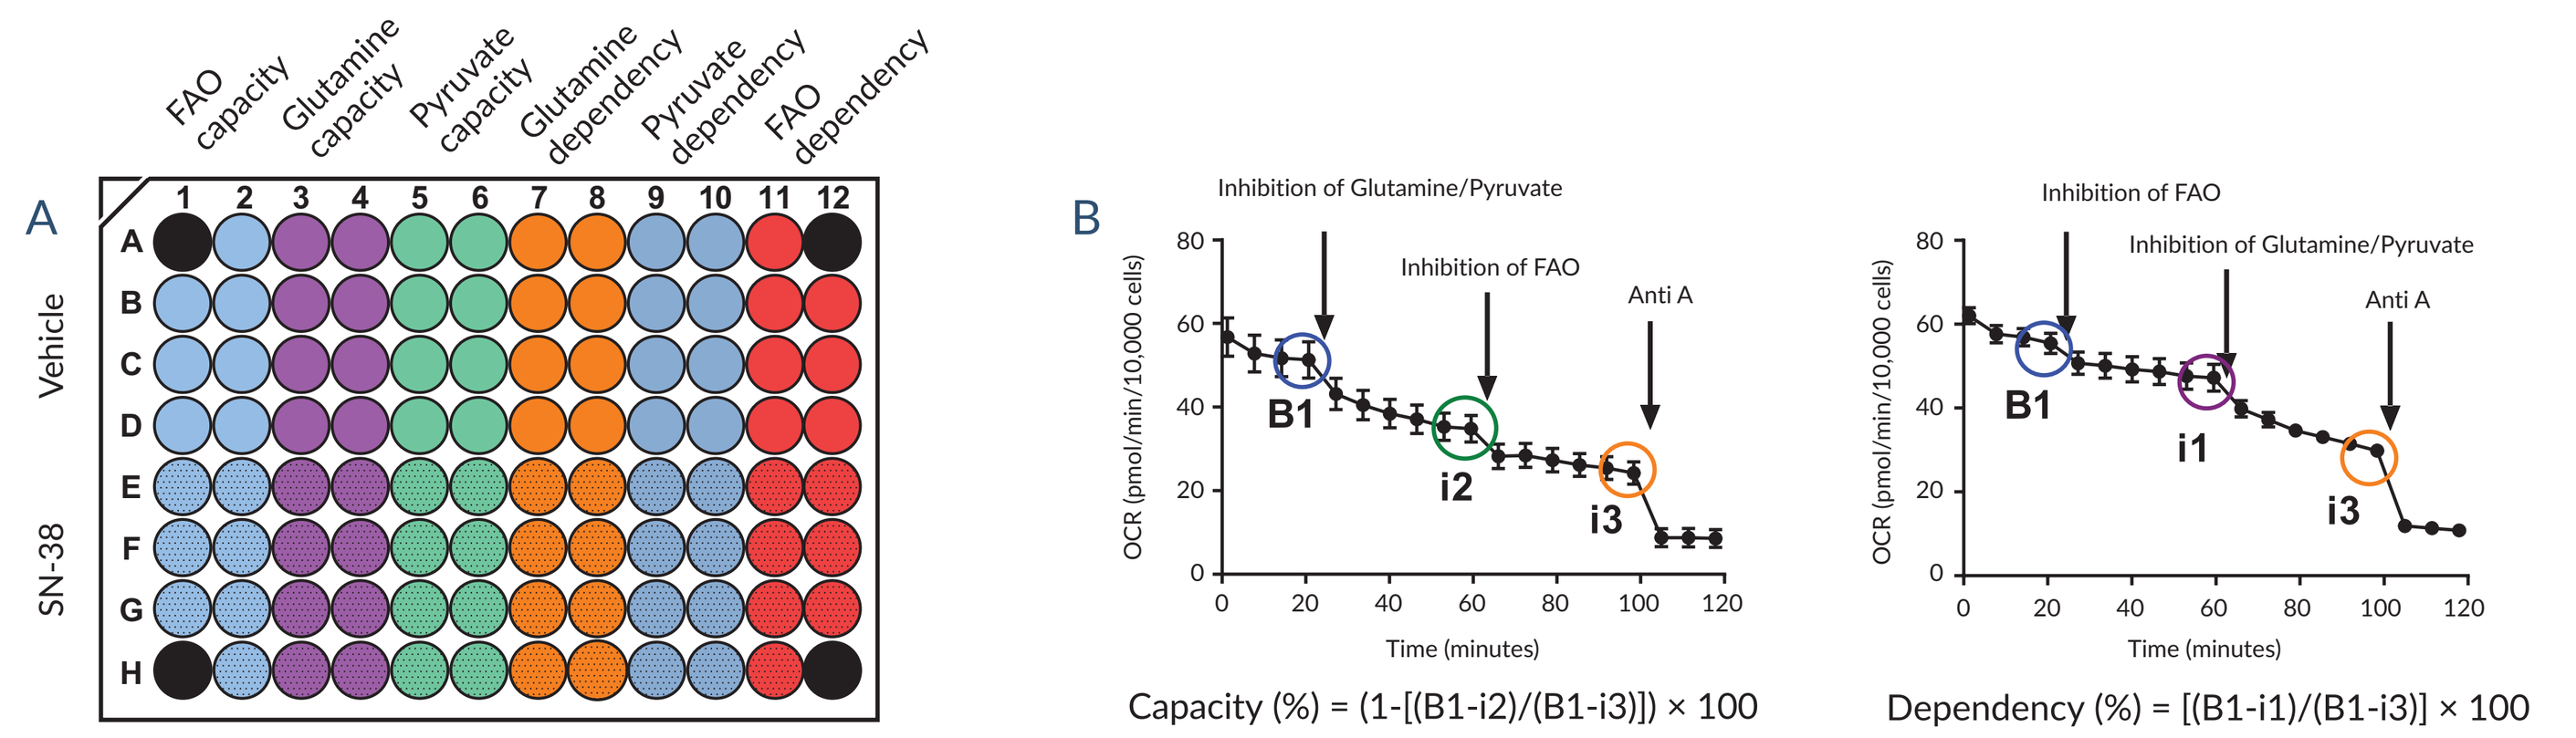

Capacity (%) = (1-[(B1-i2)/(B1-i3)]) × 100

Dependency (%) = [(B1-i1)/(B1-i3)] × 100

Figure 4. In order to determine mitochondrial fuel flexibility, cells were cultured in the presence or absence of SN-38 (30 nM) at 21% or 1% O_2. Following the treatment period, OCR was measured using the plate layout in **Figure 4A**. Capacity and dependency for FAO, glutamine, and pyruvate was determined by adding different inhibitors to inhibit transporters of each pathway. OCR values were used at points of equilibrium indicated in **Figure 4B** and used to calculate a percentage for each inhibitor combination (Vacanti, N.M., Divakaruni, A.S., Green, C.R., *et al.* Regulation of substrate utilization by the mitochondrial pyruvate carrier. *Mol. Cell* **56(3)**, 425-435 (2014)). An example trace for FAO capacity and FAO dependency for untreated cells is shown in **Figure 4B**. Inhibitors and final concentrations utilized were as follows: etomoxir (2 µM) to inhibit CPT1 (FAO), UK5099 (4 µM) to inhibit mitochondrial pyruvate oxidation, and BPTES (3 µM) to inhibit glutamine oxidation.

In HCT116 cells treated with SN-38 for 12 hours, no changes in substrate capacity are observed at 21% O_2 (**Figure 5A**). However, decreased substrate dependency is observed both for FAO and pyruvate. At 1% O_2, a decrease in glutamine capacity is observed with minimal change in glutamine dependency (**Figure 5B**). When comparing 1% to 21% O_2, an increase in pyruvate dependency is observed, which would be expected under hypoxic conditions as upregulated glycolysis results in increased pyruvate availability. In cells treated acutely with SN-38 at 21% O_2, a decrease in glutamine capacity can be observed in the presence of SN-38, as well as a slight decrease in pyruvate capacity (**Figure 5C**). A slight increase in FAO capacity in the presence of SN-38 is also observed. When looking at substrate dependency, it is clear that FAO dependency is greatly decreased in the presence of SN-38 with a slight increase in pyruvate dependency. When compared to long term treatment, these data show an effect of SN-38 on FAO (**Figure 5D**). Since the effect happens during both treatments, the data suggest it is an immediate effect and not the result of long-term changes to the cell's metabolism. As with 1% O_2 in the 12-hour SN-38 treatment, an increase in pyruvate dependency is observed. Taken together, these data show that at 21% O_2 HCT116 cells have increased dependency on FAO. This dependency is prevented by the addition of SN-38. At 1% O_2, FAO dependency is already lowered, and the addition of SN-38 has minimal effect, suggesting that at 1% O_2 this cell line can switch its metabolic phenotype to be more reliant on pyruvate. These data provide evidence that while SN-38 is capable of inhibiting FAO, HCT116 cells are less dependent on fatty acids as substrates at lower [O_2].

12-hour Treatment *Acute Treatment*

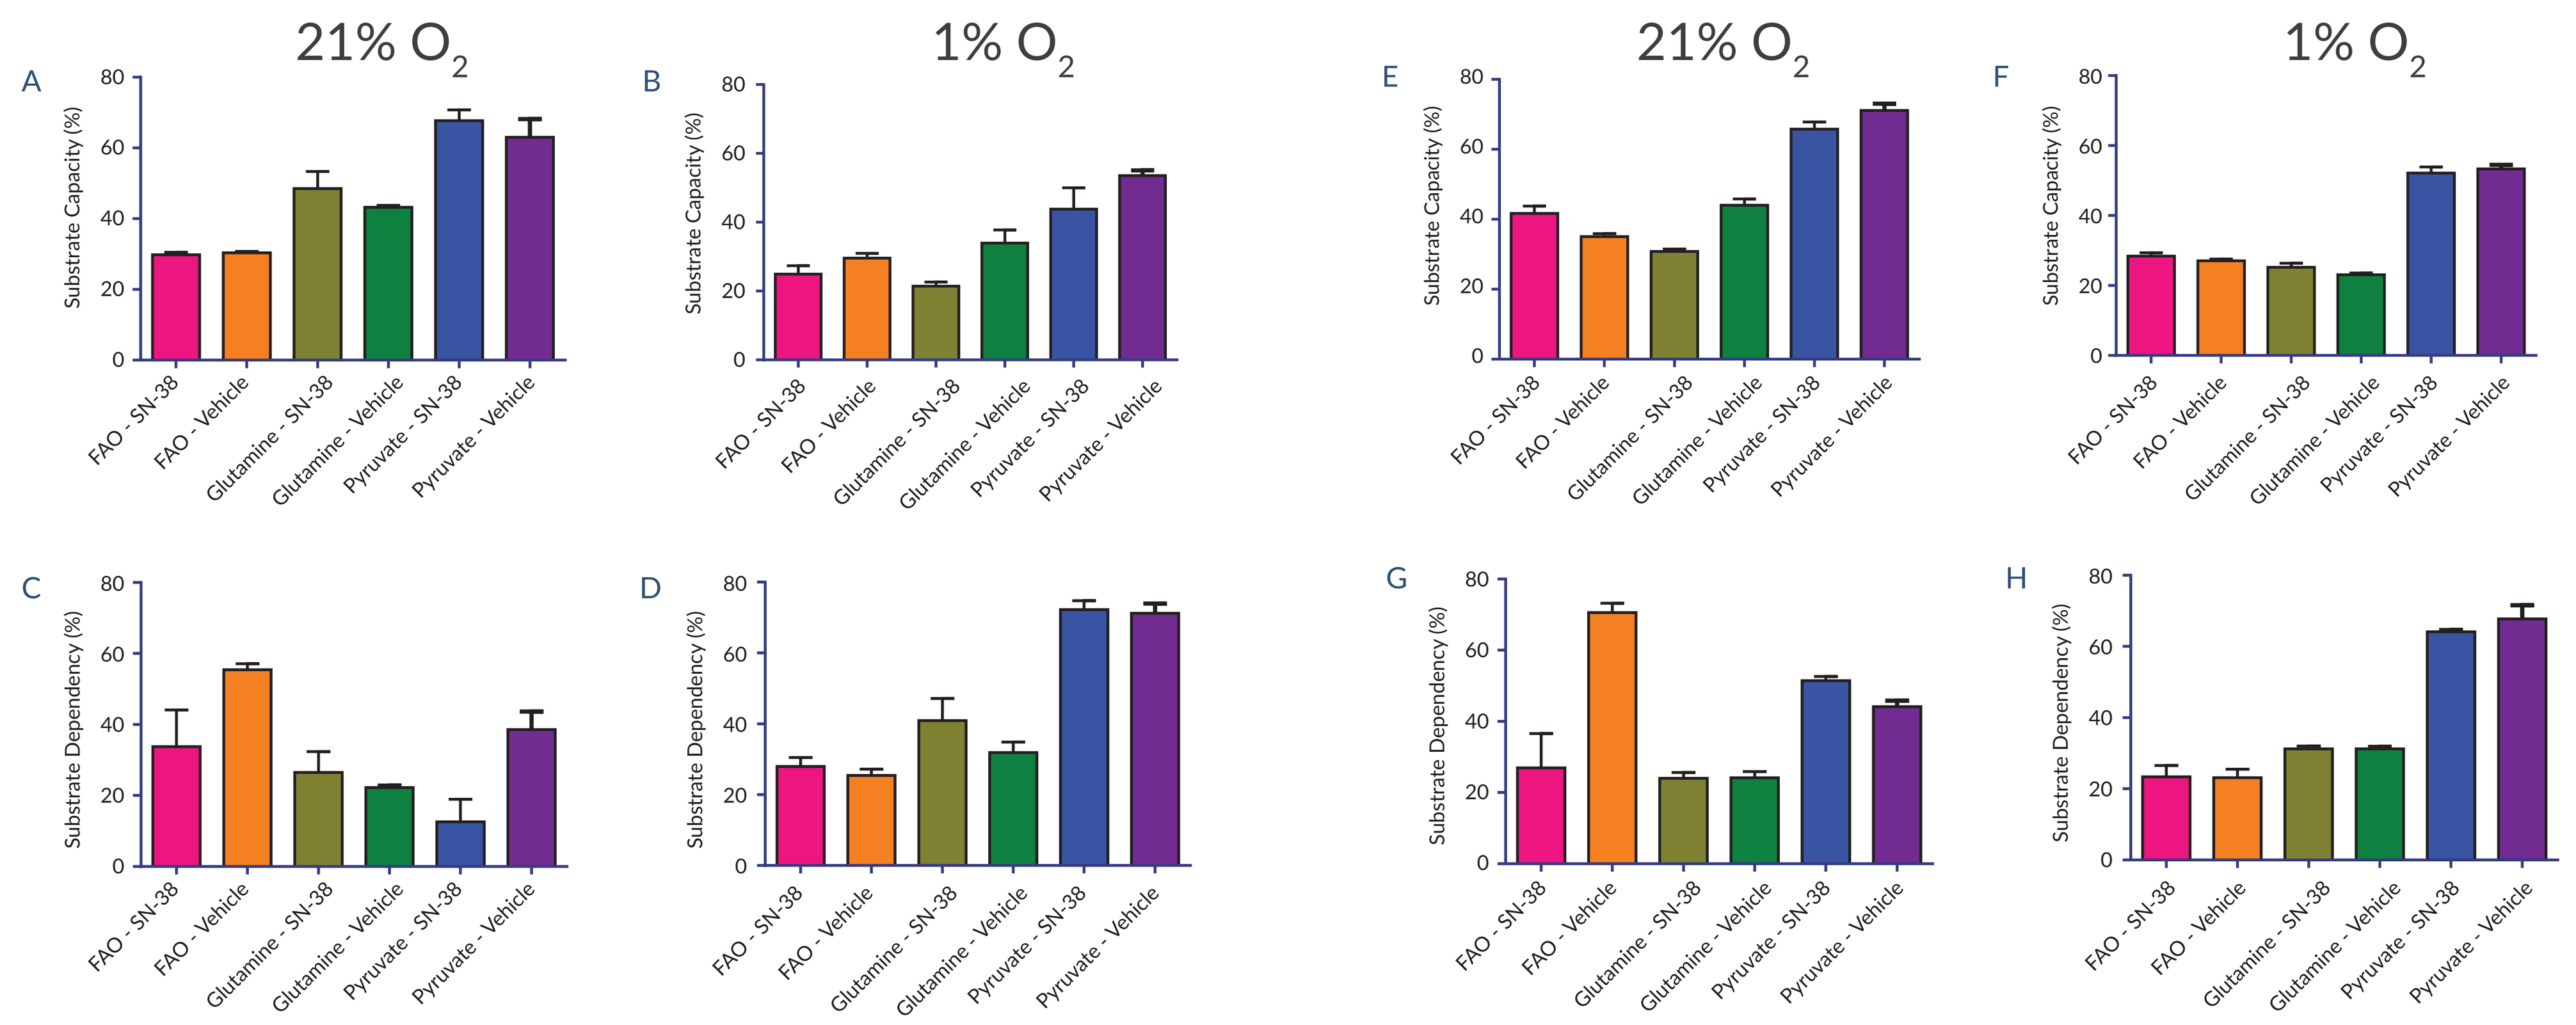

Figure 5. Substrate capacity and substrate dependency were calculated using the equations in **Figure 4B**. Calculated substrate capacities and dependencies for 12-hour treatment with 30 nM SN-38 at 21% and 1% O_2 can be found in **Figures A-D**. Calculated capacities and dependencies for acute treatment of 30 nM SN-38 for 21% and 1% O_2 can be found in **Figures E-H**. Data are presented as means ± standard error.

Differentiation of MDSCs and Measurement of Metabolic Phenotype

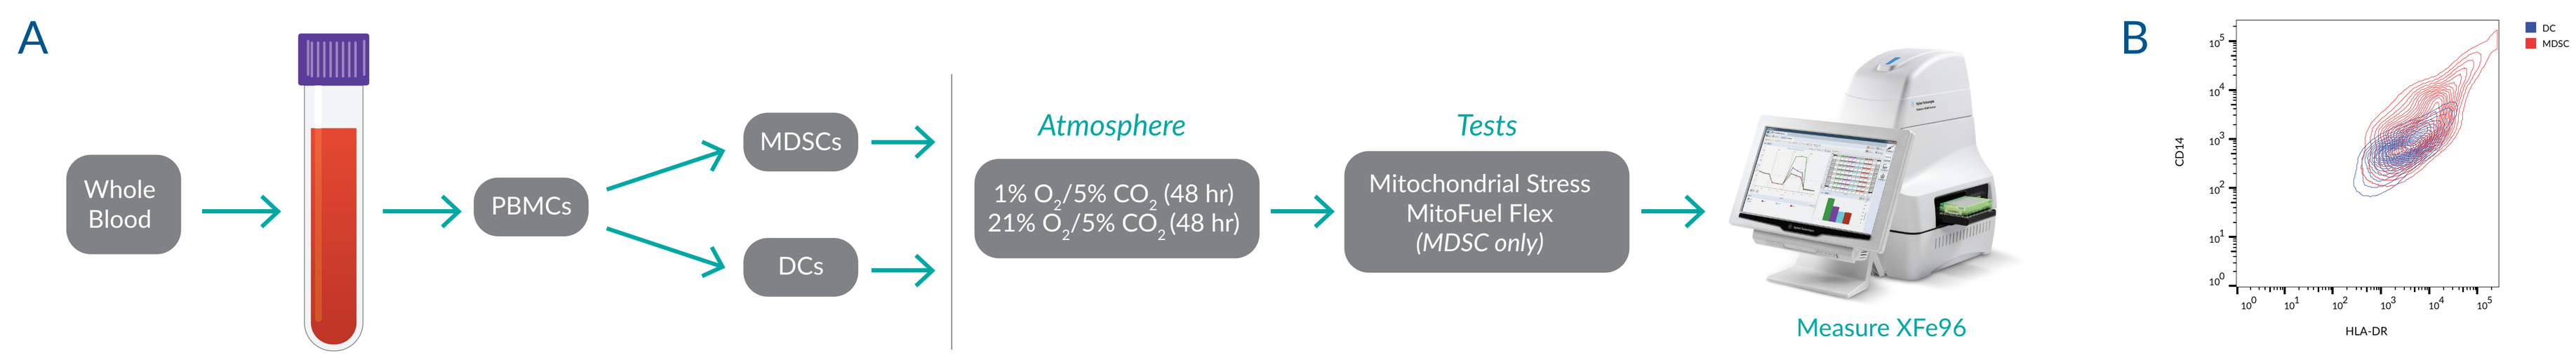
Whole Blood
PBMCs
MDSCs
DCs
Atmosphere
1% O_2/5% CO_2 (48 hr)
21% O_2/5% CO_2 (48 hr)
Tests
Mitochondrial Stress
MitoFuel Flex
(MDSC only)
Measure XFe96

Figure 6. A. Peripheral blood mononuclear cells (PBMCs) isolated from whole blood monocytes were isolated from PBMCs immediately prior to isolation of untouched monocytes (Dynabeads® Untouched™ Human Monocytes Kit (ThermoFisher)). Following isolation, monocytes were stimulated with IL-4 and GM-CSF to facilitate differentiation into dendritic cells (DCs), or IL-4 and GM-CSF plus 1 µM prostaglandin E_2 to facilitate differentiation into MDSCs **B.** Following differentiation, cells were stained for CD14 and HLA-DR to determine characteristics of the two cell types.

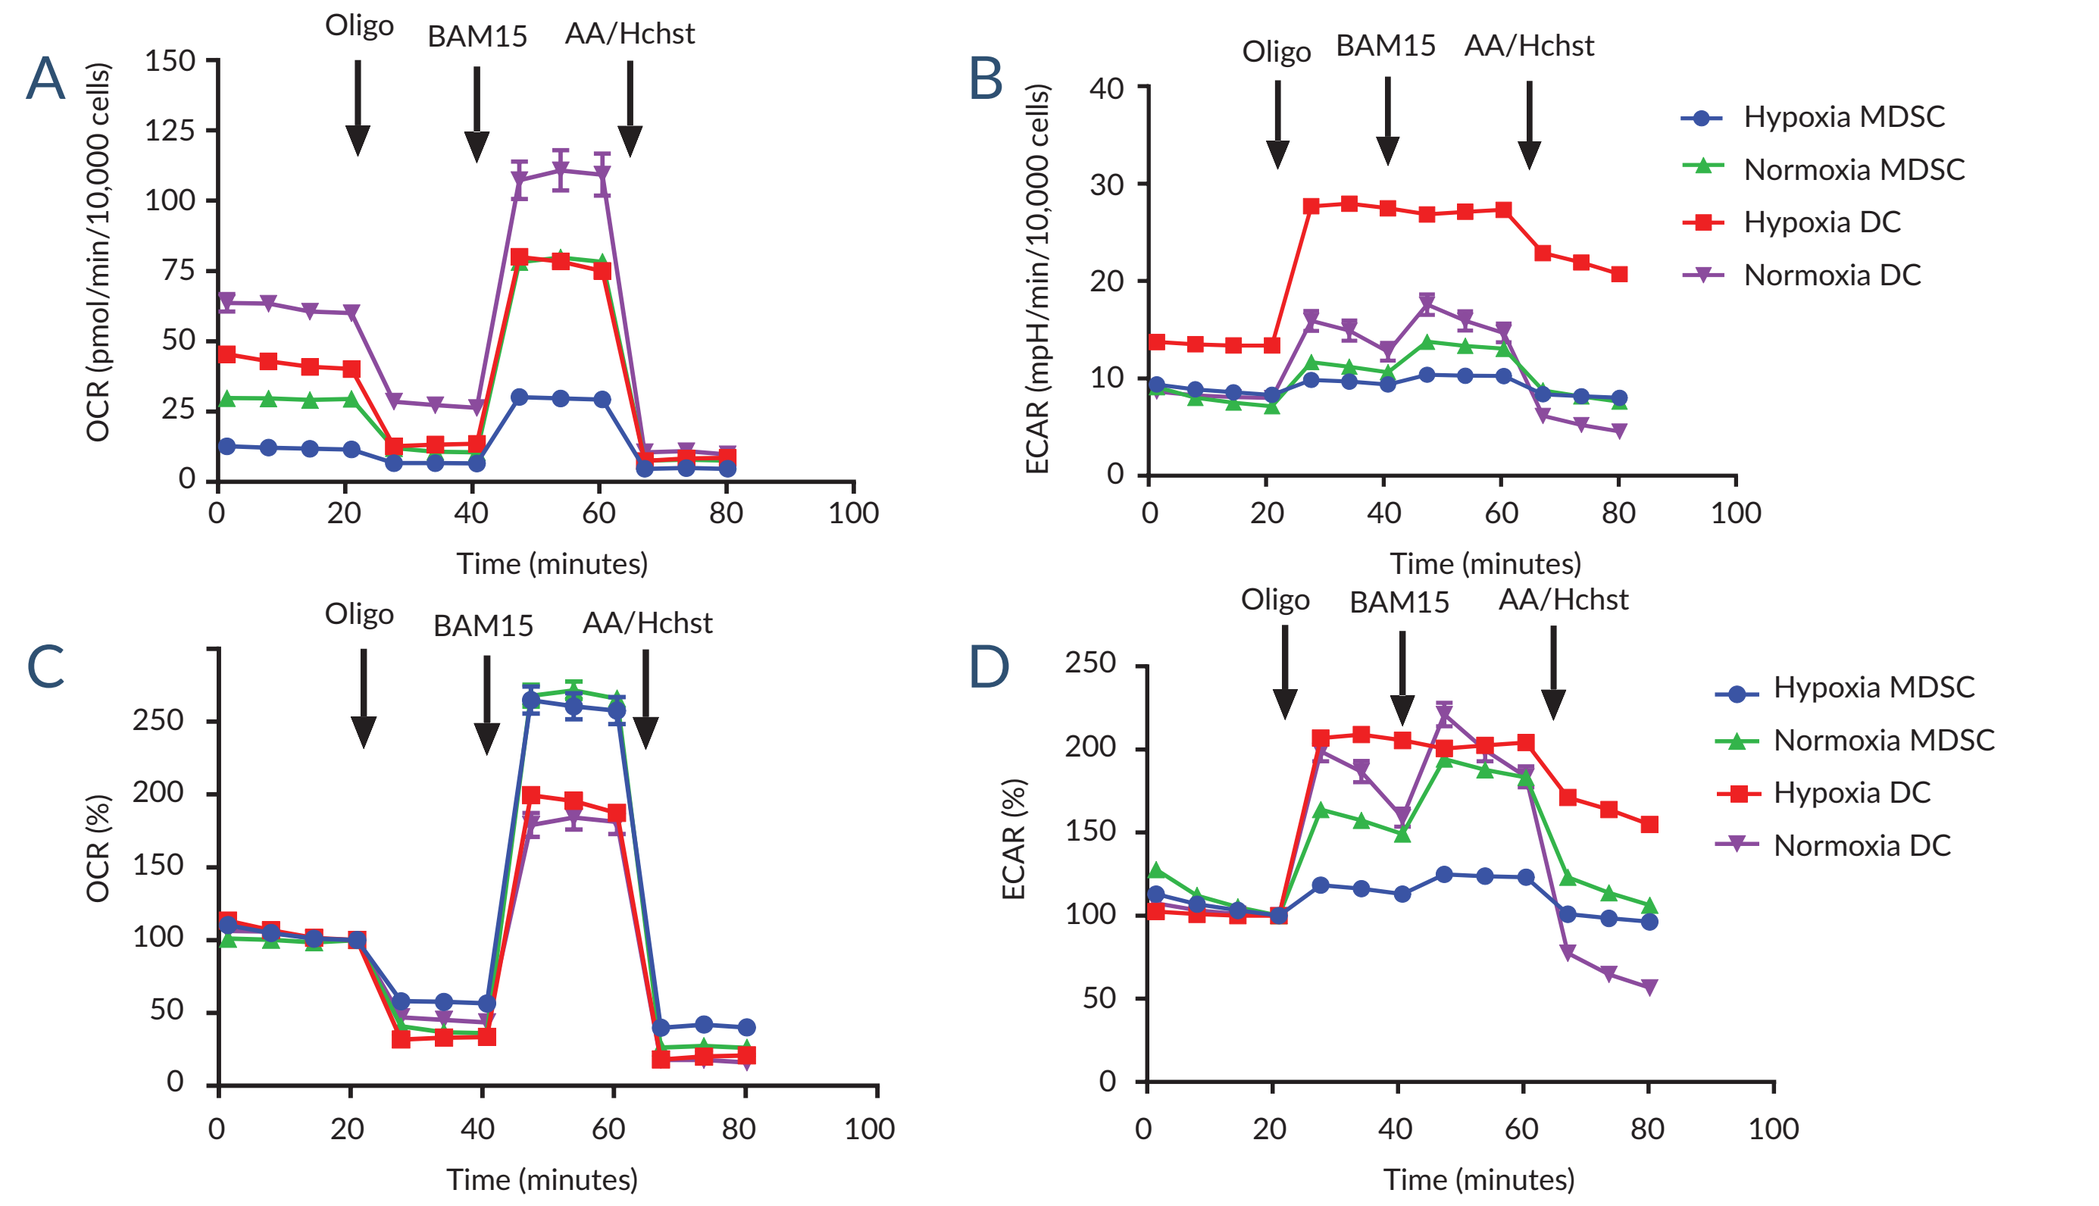

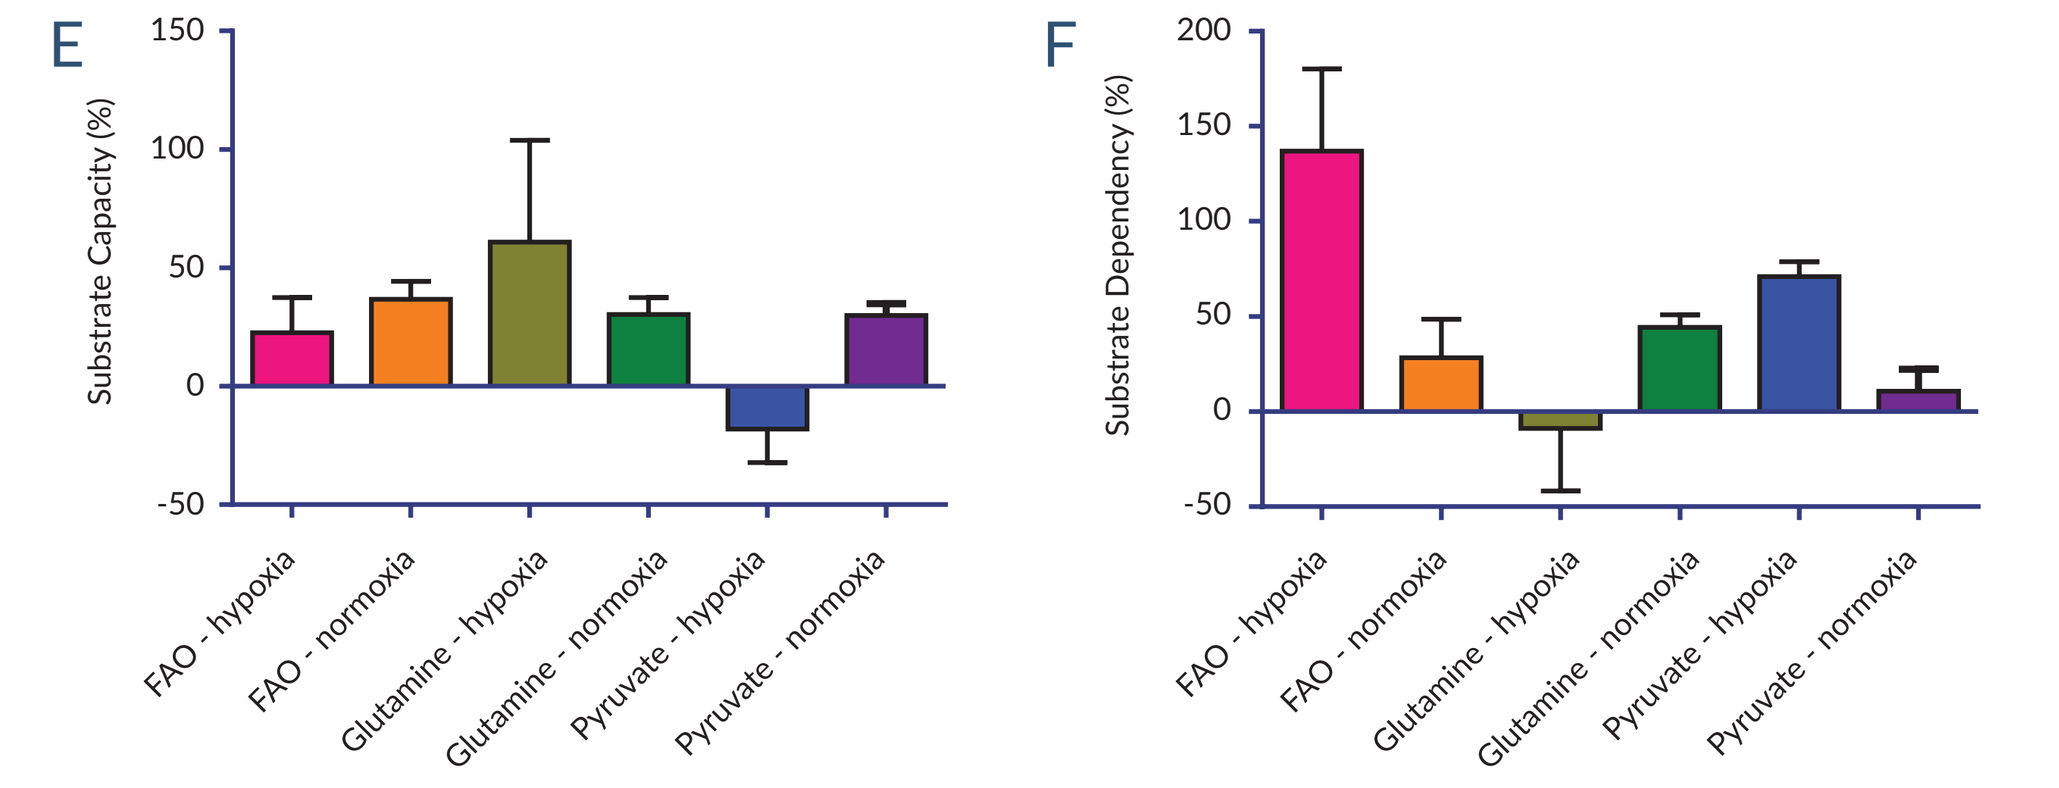

Figure 7. A-B. MDSCs and DCs that were differentiated at 21% and 1% O_2 were subjected to a mitochondrial stress test as detailed in Figures 1 and 2. **C-D.** Data were normalized to baseline to illustrate % change in OCR independently of cell number. **E-F.** MDSCs differentiated at 21% and 1% O_2 were subjected to a mitochondrial fuel flex test as described in **Figure 4.**

Since MDSCs are thought to occupy the TME—potentially suppressing T cell function, metabolic function tests were performed on MDSCs and dendritic cells (DCs) cultured at 21% and 1% O_2 during the last two days of differentiation. These tests were performed in order to better understand how the TME can affect metabolic phenotype. **Figure 7A** and **7B** shows a mitochondrial stress test performed on MDCSs and DCs cultured as described above. OCR values normalized to cell number show DCs differentiated at 21% O_2 are expectedly more aerobic than those cultured at 1% O_2, possessing a higher basal OCR (**Figure 7A**). MDSCs behave in a similar manner, with those differentiated at 21% O_2 having a higher basal OCR compared to those differentiated under 1% O_2. ECAR values for DCs are consistent with OCR values in that cells differentiated under 1% O_2 have higher ECAR values, showing higher rates of glycolysis (**Figure 7B**). Interestingly, MDSCs do not appear to follow this pattern. Upon normalization to baseline, it becomes apparent that MDSCs, regardless of O_2 concentration, have an increased response to uncoupler (BAM15) when compared to DCs, indicating a different metabolic phenotype (**Figure 7C**). To further investigate this, a mitochondrial fuel flex test was performed on MDSCs differentiated at 1% and 21% O_2. The data show that MDSCs that were differentiated at 1% have an increased dependency on FAO, suggesting that lower [O_2] has the potential to alter the metabolic phenotype of MDSCs.

Future directions include measuring metabolic phenotype in MDSCs differentiated at 21% and 1% O_2 in the presence and absence of SN-38 to determine if the observed effect on metabolic phenotype can be ablated.